P1505

A Multi-Site Study Comparing a Commercially Prepared Dried MIC Susceptibility System to the CLSI/ISO Broth Microdilution Method for Sulbactam/durlobactam (SE4) using *Acinetobacter* spp.

D. Staats, T. Lewis, N. Holliday, C. Knapp, S. Killian, B. Olson, C.L. Pike, E.K. Higdon, R.J. Schoone, T. Fritsche, A. Gattis, N. Waugh, K. Doing, P. von Stein, A. Goer, K. Knight, D. Butler, **D. Paisey**

ThermoFisher SCIENTIFIC

Abstract

Purpose: Sulbactam-durlobactam is a novel β-lactam/β-lactamase inhibitor combination targeted to treat carbapenem-resistant *Acinetobacter baumannii*. A 4-site evaluation was performed to determine the accuracy and reproducibility of SE4 susceptibility testing against *Acinetobacter* spp. using the Thermo ScientificTM SensititreTM dried MIC susceptibility system compared with the CLSI (M07, M100)/ ISO 20776-1, ISO 20776-2 (CLSI/ISO) reference broth microdilution method (BMD).

Methods: The Sensititre 18-24 Hour MIC or Breakpoint Susceptibility System with SE4 in the dilution range of 0.015/4-32/4 µg/ml was used to test 297 recent clinical and challenge isolates and 10 reproducibility isolates. Microorganisms tested included 86 Acinetobacter spp., and 210 *Acinetobacter baumannii*. The Sensititre dried MIC susceptibility system was inoculated per manufacturers' instructions. BMD was performed per CLSI/ISO guidelines. Recommended CLSI quality control (QC) organisms were tested daily and all results were within the published QC ranges.

Results: Comparisons of the indicated microorganisms' MIC results on the Sensititre system to the CLSI/ISO BMD for both automated and manual reads resulted in a calculated essential agreement of 96.5% for manual and 95.5% automated read methods (EA; +/- 1 $\log_2$ dilution) for sulbactam-durlobactam. Categorical Agreement was shown to be 97.2% and 96.9% for automated and manual read methods respectively. Overall agreement for the reproducibility (+/- 1 $\log_2$ dilution of the modal MIC) using automated and manual reads was 99.4% and 97.8%, respectively.

Conclusions: The Sensititre susceptibility system demonstrates an equivalent level of performance compared to the CLSI/ISO BMD method when testing sulbactam-durlobactam against *Acinetobacter* spp. The high level of agreement obtained by the Sensititre susceptibility system and the CLSI/ISO BMD method suggests that this is an acceptable method for susceptibility testing of sulbactam-durlobactam.

Introduction

Sulbactam-durlobactam (Innoviva Specialty Therapeutics) was developed for the treatment of hospital-acquired bacterial pneumonia and ventilator-associated bacterial pneumonia (HABP/VABP) caused by susceptible isolates of *Acinetobacter baumannii-calcoaceticus complex*. Sulbactam-durlobactam is a combination of sulbactam, which inhibits cell wall synthesis by inhibiting PBP-1 and PBP-3, and durlobactam used to prevent the hydrolysis of Sulbactam. Sulbactam-durlobactam is included in the IDSA 2024 guidance for Gram-Negative Infections.

A four-site evaluation was performed to determine the accuracy and reproducibility of SE4 susceptibility testing using the Sensititre™ dried MIC susceptibility system (Thermo Fisher Scientific, Cleveland, OH) compared with the CLSI M07/ISO 20776-1/ISO 20776-2 (CLSI/ISO) reference broth microdilution method (BMD). Both auto (Optiread™) and manual read methodologies were employed.

Materials and methods

- Sensititre dried MIC susceptibility plates (Thermo Fisher Scientific, East Grinstead, UK) (**Fig. 1**)
- CLSI M07 reference broth microdilution method (BMD) plates (Thermo Fisher Scientific, Cleveland, OH)
- 287 clinical and challenge isolates (**Table 1**)
- CLSI recommended quality control strain (**Table 2**)
- 10 reproducibility isolates (**Table 1, Table 5**)

Materials and methods (cont.)

Table 1. Organisms Tested

Species	Isolates Tested
Acinetobacter baumannii – calcoaceticus complex	297

All 287 clinical and challenge isolates were tested against SE4 (0.015/4-32/4µg/mL) on both the Sensititre dried plates and the frozen reference BMD plates. Dried plates were tested according to manufacturer's instructions. Frozen reference plates were tested according to CLSI M07. Dried plates were read manually using the Sensititre™ Optiread and Vizion and frozen plates were read using a manual mirror box.

Figure 1. Sensititre Dried MIC Susceptibility Plates

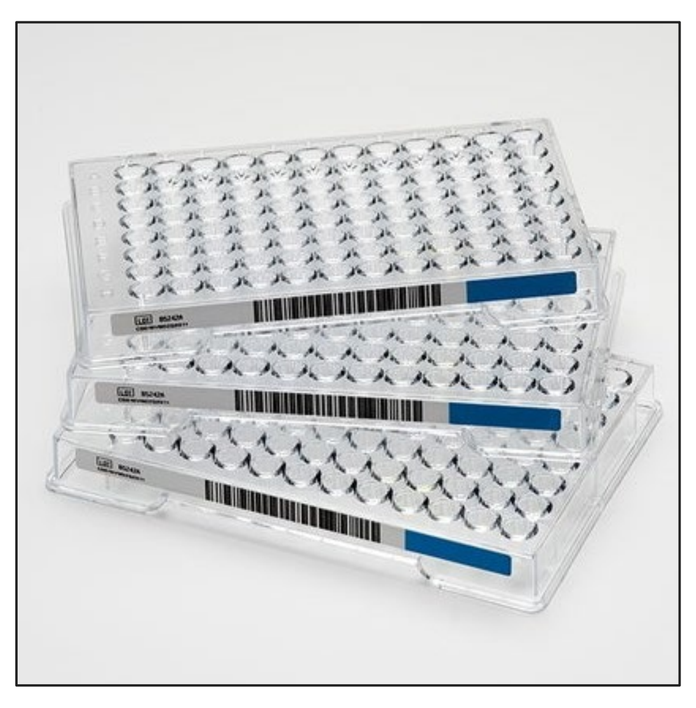

Recommended CLSI quality control organisms were tested daily and were within the expected ranges. Colony counts were performed daily for each QC strain to verify the isolates correct inoculum concentration.

Table 2. Quality Control Strains

Quality Control Strain	CLSI QC Ranges (µg/ml) Sulbactam-durlobactam
Acinetobacter baumannii NCTC 13304	0.5/4 – 2/4

Results

Agreement:

Essential agreement for SE4 on the Sensititre 18 – 24 hour susceptibility plate compared to the frozen reference microdilution plate was calculated using the ±1 $\log_2$ dilution standard. Categorical agreement was calculated utilizing the CLSI breakpoints of ≤4/4, 8/4, ≥16/4.

The essential agreement of SE4 after initial testing was 95.5% and 96.5% when testing automated and manual reads respectively. Categorically, agreement was determined to be 97.2% and 96.9% respectively (Not Presented). Total biases were within the ± 30% limits as set in ISO (Table 3 and 4).

Reproducibility:

Reproducibility for SE4 on the Sensititre 18 – 24 hour susceptibility plate was determined by comparing individual results to the overall modal MIC determined at 4 sites. Testing was conducted on the test system only in triplicate over 3 days. Overall, both read methods showed acceptable reproducibility as shown in Table 5.

Conclusions

This study validated that the Sensititre susceptibility system demonstrates an equivalent level of performance compared to the CLSI/ISO reference broth microdilution plate when testing SE4 against clinically relevant *Acinetobacter baumannii-calcoaceticus* complex isolates. This study suggests that the Sensititre system is an acceptable method for susceptibility testing of SE4 according to both the CLSI and ISO testing methodology

Table 3. Summary Data and % Essential Agreement of *Acinetobacter* spp. isolates autoread on the Sensititre 18 – 24 Hour Susceptibility Test System

Sulbactam-durlobactam							
Organism Group	Total of all Isolates	Total Evaluable Isolates	Essential Agreement of Total	Essential Agreement of Evaluable	% Essential Agreement of Total	% Essential Agreement of Evaluable	Total Bias
Acinetobacter baumannii – calcoaceticus complex	287	281	274	268	95.5%	95.4%	13.0 %

Table 4. Summary Data and % Essential Agreement of *Acinetobacter* spp. isolates read manually on the Sensititre 18 – 24 Hour Susceptibility Test System

Sulbactam-durlobactam							
Organism Group	Total of all Isolates	Total Evaluable Isolates	Essential Agreement of Total	Essential Agreement of Evaluable	% Essential Agreement of Total	% Essential Agreement of Evaluable	Total Bias
Acinetobacter baumannii – calcoaceticus complex	286*	279	276	269	96.5%	96.4%	2.6%

*One isolate was not read on the Vizion system

Table 5. Reproducibility Data for Automated (Left) and Manual (Right) read methods for sulbactam-durlobactam on the Sensititre 18-24 Susceptibility Plate

SE4	Difference in the number of wells between test result and test mode (Auto Read)						
	OFF-Scale	≤-2	-1	0	+1	≥+2	OFF-Scale
***Acinetobacter baumannii – calcoaceticus* complex**				32	4		
***Acinetobacter baumannii – calcoaceticus* complex**				31	5		
***Acinetobacter baumannii – calcoaceticus* complex**				29	5	**2**	
***Acinetobacter baumannii – calcoaceticus* complex**			2	32	2		
***Acinetobacter baumannii – calcoaceticus* complex**				36			
***Acinetobacter baumannii – calcoaceticus* complex**				21	15		
***Acinetobacter baumannii – calcoaceticus* complex**				24	12		
***Acinetobacter baumannii – calcoaceticus* complex**			2	33	1		
***Acinetobacter baumannii – calcoaceticus* complex**				35	1		
***Acinetobacter baumannii – calcoaceticus* complex**			12	23			
TOTAL	**0**	**0**	**17**	**296**	**45**	**2**	**0**
Reproducibility Essential Agreement	371/378 = 99.4%						

SE4	Difference in the number of wells between test result and test mode (Manual Read)						
	OFF-Scale	≤-2	-1	0	+1	≥+2	OFF-Scale
***Acinetobacter baumannii – calcoaceticus* complex**		2	16	17	1		
***Acinetobacter baumannii – calcoaceticus* complex**				30	6		
***Acinetobacter baumannii – calcoaceticus* complex**			1	27	3	**5**	
***Acinetobacter baumannii – calcoaceticus* complex**			4	29	3		
***Acinetobacter baumannii – calcoaceticus* complex**				36			
***Acinetobacter baumannii – calcoaceticus* complex**			2	27	7		
***Acinetobacter baumannii – calcoaceticus* complex**			6	21	9		
***Acinetobacter baumannii – calcoaceticus* complex**			2	29	5		
***Acinetobacter baumannii – calcoaceticus* complex**			10	25		1	
***Acinetobacter baumannii – calcoaceticus* complex**			17	19			
TOTAL	**0**	**2**	**58**	**260**	**34**	**6**	**0**
Reproducibility Essential Agreement	352/360 = 97.8%						

Science at a scan
Scan the QR code on the right with your mobile device to download this and many more scientific posters.

Learn more at thermofisher.com/sensititre