Population Pharmacokinetics Analysis of Eravacycline to Support Dose Selection for a Trial in Pediatric Patients Aged 8 to Less Than 18 Years with Complicated Intra-Abdominal Infection

Poster P-443

Claudia Jomphe[1], Nathalie H. Gosselin[1], Angela Tanudra[2], Khurram Rana[2], Drew Lewis[2], and Kajal B. Larson[2]

[1]Certara Drug Development Solutions, Princeton, NJ, USA; [2]Innoviva Specialty Therapeutics, Inc. Waltham, MA USA

Innoviva Specialty Therapeutics, Inc.
930 Winter Street Suite 1500
Waltham, MA 02451
Kajal.Larson@istx.com

INNOVIVA Specialty Therapeutics™

Background

Eravacycline (ERV) is a synthetic fluorocycline antibiotic within the tetracycline class, exhibiting broad-spectrum activity against a range of Gram-positive and Gram-negative pathogens, including multidrug-resistant organisms. ERV is currently approved for the treatment of complicated intra-abdominal infections (cIAI) in adults, based on its demonstrated efficacy and safety profile in pivotal clinical trials.

To support pediatric development, a Phase 1, open-label, single-dose pharmacokinetic (PK) trial with ERV was conducted in children aged 8 to <18 years with suspected or confirmed bacterial infections (ClinicalTrials.gov Identifier: NCT03696550). These patients were receiving systemic antibiotic therapy other than ERV, at the time of enrollment. The primary objectives of the trial were to characterize the PK parameters of ERV following a single 1-hour intravenous (IV) infusion, and to assess its safety and tolerability in the pediatric population. Subjects were stratified into two age-based cohorts:

- Cohort 1: 12 to <18 years (N=9) and received a single dose of 1.50 mg/kg ERV
- Cohort 2: 8 to <12 years (N=10) and received a single dose of 1.75 mg/kg ERV

The PK data obtained from the Phase 1 trial were integrated into a population PK model. This model enabled simulation-based dose optimization and informed the design of a Phase 2 clinical trial aimed at evaluating ERV in pediatric patients with cIAI (NCT06794541).

Methods

Phase 1 trial

- Safety assessments included monitoring of vital signs, electrocardiogram parameters, clinical safety laboratory tests, physical examinations and adverse events monitoring
- Blood samples for PK analysis were collected at the following timepoints:
 - Pre-dose (within 30 minutes before infusion)
 - 5–10 minutes post-infusion
 - 0.5–1 hour, 2.5–3.5 hours, and 4–5 hours post-infusion
 - 24 hours post-infusion (aligned with safety assessments)
- PK parameters were derived using population PK modeling, with individual exposures derived from model-predicted concentration profiles using actual dosing regimen and posterior Bayesian estimates

Population PK model for Phase 2 trial

- A PPK model was developed using data from 12 trials of ERV in adults and the Phase 1 pediatric trial
- Model-based simulations, using a virtual pediatric population (250 males and 250 females) with age-weight matched information, were conducted for two age groups: 8 to < 12 and 12 to < 18 years of age
- For adults, cIAI patient characteristics were resampled from the current PK population
- Dosing regimens were evaluated based on their ability to yield pediatric exposures that:
 - approximated the geometric mean AUC_{0-24} in cIAI patients (12,146 h•ng/mL) and
 - did not exceed the maximum exposures previously demonstrated to be safe in adults (C_{max} of 6,400 ng/mL)

Table 1. Summary of Predicted Noncompartmental Eravacycline PK Parameters in Children, Adolescents, and Adults with Infection

	Pediatric Patients with Bacterial Infection		Adult Patients with cIAI	
	1.5 mg/kg (adolescents, N= 9)	1.75 mg/kg (children, N= 10)	1 mg/kg (N=555)	1.5 mg/kg (N=52)
C_{max} (ng/mL)				
Median [Min, Max]	1856 [1030, 2422]	1477 [1053, 2272]	994 [191, 7301]	1546 [305, 2939]
Geometric Mean (%CV)	1666 (27.7%)	1440 (22.5%)	955 (47.1)	1373 (43.8%)
AUC_{0-24} (h•ng/mL)				
Median [Min, Max]	4477 [2229, 5900]	5024 [3432, 5496]	4050 [1361, 12856]	4938 [2228, 7800]
Geometric Mean (%CV)	4307 (32.3%)	4838 (14.2%)	4055 (30.7%)	4985 (27.1%)
Half-life (hours)				
Median [Min, Max]	11.6 [8.97, 13.8]	11.8 [10.6, 14.0]	14.0 [8.40, 105]	14.1 [7.89, 34.4]
Geometric Mean (%CV)	11.5 (12.3%)	11.8 (7.28%)	14.4 (26.0%)	14.5 (28.1%)

Abbreviations: %CV, percent coefficient of variation; AUC_{0-24}, area under the curve from time 0 to 24 hours, cIAI, complicated intra-abdominal infection; C_{max}, maximum plasma concentration; max, maximum; min, minimum; PK, pharmacokinetic

Results

Phase 1 trial

Baseline Characteristics

The Cohort 1 population (9 adolescents) had a median age of 15 years (range: 12 to 17 years) and body weight of 60.6 kg (range: 32.4 to 77.7 kg). The proportions of female and male adolescents were 33.3% and 66.7%, respectively.

The Cohort 2 population (10 children) had a median age of 11 years (range: 9 to 11 years) and body weight of 36.5 kg (range: 28.5 to 66.2 kg). The proportions of female and male children were 20.0% and 80.0%, respectively.

PK

A 3-compartment model with linear elimination adequately characterized the observed plasma ERV concentration-time profiles and was considered robust for interim and final analyses of the PK for the Phase 1 trial.

- Table 1 shows the predicted noncompartmental PK parameters of ERV in children, adolescents, and adults
- Of note, the model determined the PK parameters based on the Phase 1 pediatric trial, in which no cIAI pediatric patients were enrolled

Safety

ERV was considered safe and generally well tolerated by the trial. A summary of TEAEs is shown in Table 2.

- The majority of TEAEs were mild or moderate in severity and considered possibly or probably related to study drug
- Severe TEAEs were reported in 2 subjects in Cohort 1 (severe anaphylactic reaction and severe pleural effusion)
- There were no deaths and no TEAEs that led to early treatment or study discontinuation
- Two serious adverse events (SAEs) were reported in 2 subjects during the study (pleural effusion and gastroenteritis salmonella)
 - The event of gastroenteritis salmonella was not considered a TEAE but was reported as an SAE

Table 2. Summary of Treatment-Emergent Adverse Events (Safety Population) in the Phase 1 trial

	Cohort 1 (N=9)	Cohort 2 (N=10)	Total (N=19)
Total Number of TEAEs	19	6	25
Number of Subjects with at Least One TEAE	7 (77.8%)	5 (50.0%)	12 (63.2%)
Gastrointestinal disorders	5 (55.6%)	1 (10.0%)	6 (31.6%)
Nausea	5 (55.6%)	0	5 (26.3%)
Vomiting	2 (22.2%)	1 (10.0%)	3 (15.8%)
Constipation	1 (11.1%)	0	1 (5.3%)
Tongue ulceration	1 (11.1%)	0	1 (5.3%)
Skin and subcutaneous tissue disorders	3 (33.3%)	2 (20.0%)	5 (26.3%)
Hyperhidrosis	1 (11.1%)	1 (10.0%)	2 (10.5%)
Dermatitis contact	1 (11.1%)	0	1 (5.3%)
Pruritus	0	1 (10.0%)	1 (5.3%)
Rash	1 (11.1%)	0	1 (5.3%)
Nervous system disorders	2 (22.2%)	1 (10.0%)	3 (15.8%)
Headache	2 (22.2%)	1 (10.0%)	3 (15.8%)
Psychiatric disorders	1 (11.1%)	1 (10.0%)	2 (10.5%)
Adjustment disorder with anxiety	0	1 (10.0%)	1 (5.3%)
Insomnia	1 (11.1%)	0	1 (5.3%)
Respiratory, thoracic and mediastinal disorders	2 (22.2%)	0	2 (10.5%)
Nasal congestion	1 (11.1%)	0	1 (5.3%)
Pleural effusion	1 (11.1%)	0	1 (5.3%)
General disorders and administration site conditions	1 (11.1%)	0	1 (5.3%)
Infusion site pain	1 (11.1%)	0	1 (5.3%)
Immune system disorders	1 (11.1%)	0	1 (5.3%)
Anaphylactic reaction	1 (11.1%)	0	1 (5.3%)
Metabolism and nutrition disorders	0	1 (10.0%)	1 (5.3%)
Decreased appetite	0	1 (10.0%)	1 (5.3%)

Abbreviations: TEAE, treatment-emergent adverse event;
Note: A TEAE was defined as any AE that occurred after administration of ERV or any event that was present at baseline but worsened in intensity or was subsequently considered drug-related by the Investigator. At each level of subject summarization, a subject was counted once if 1 or more events were reported for that subject. Percentages were based on the number of subjects in the safety population per cohort and Total. Adverse events were coded using MedDRA Version 24.0.

Dose selection for Phase 2 trial

The population PK model was re-evaluated to better predict exposures following multiple IV doses in pediatric patients with cIAI

A 3-compartment model with linear elimination adequately characterized ERV data.

The model included allometric functions on clearances and on volumes of distribution, severe hepatic impairment on clearance(CL), cytochrome P450 inhibition and induction on CL and central volume of distribution (V1), and infection types on V1 (cIAI) vs complicated urinary tract infection vs other infections

- Figure 1 shows the visual predictive check and indicate that the model was adequate to capture pediatric concentration
- Table 3 shows the typical population PK parameters of ERV
- Figure 2 shows the predicted steady-state exposures by weight group in children and adolescents with cIAI
- Table 4 shows the predicted exposures at doses anticipated to be safe and efficacious in pediatric patients with cIAI

Figure 2. Predicted Steady-State Exposures by Weight Groups in children (A) and adolescents (B) with cIAI

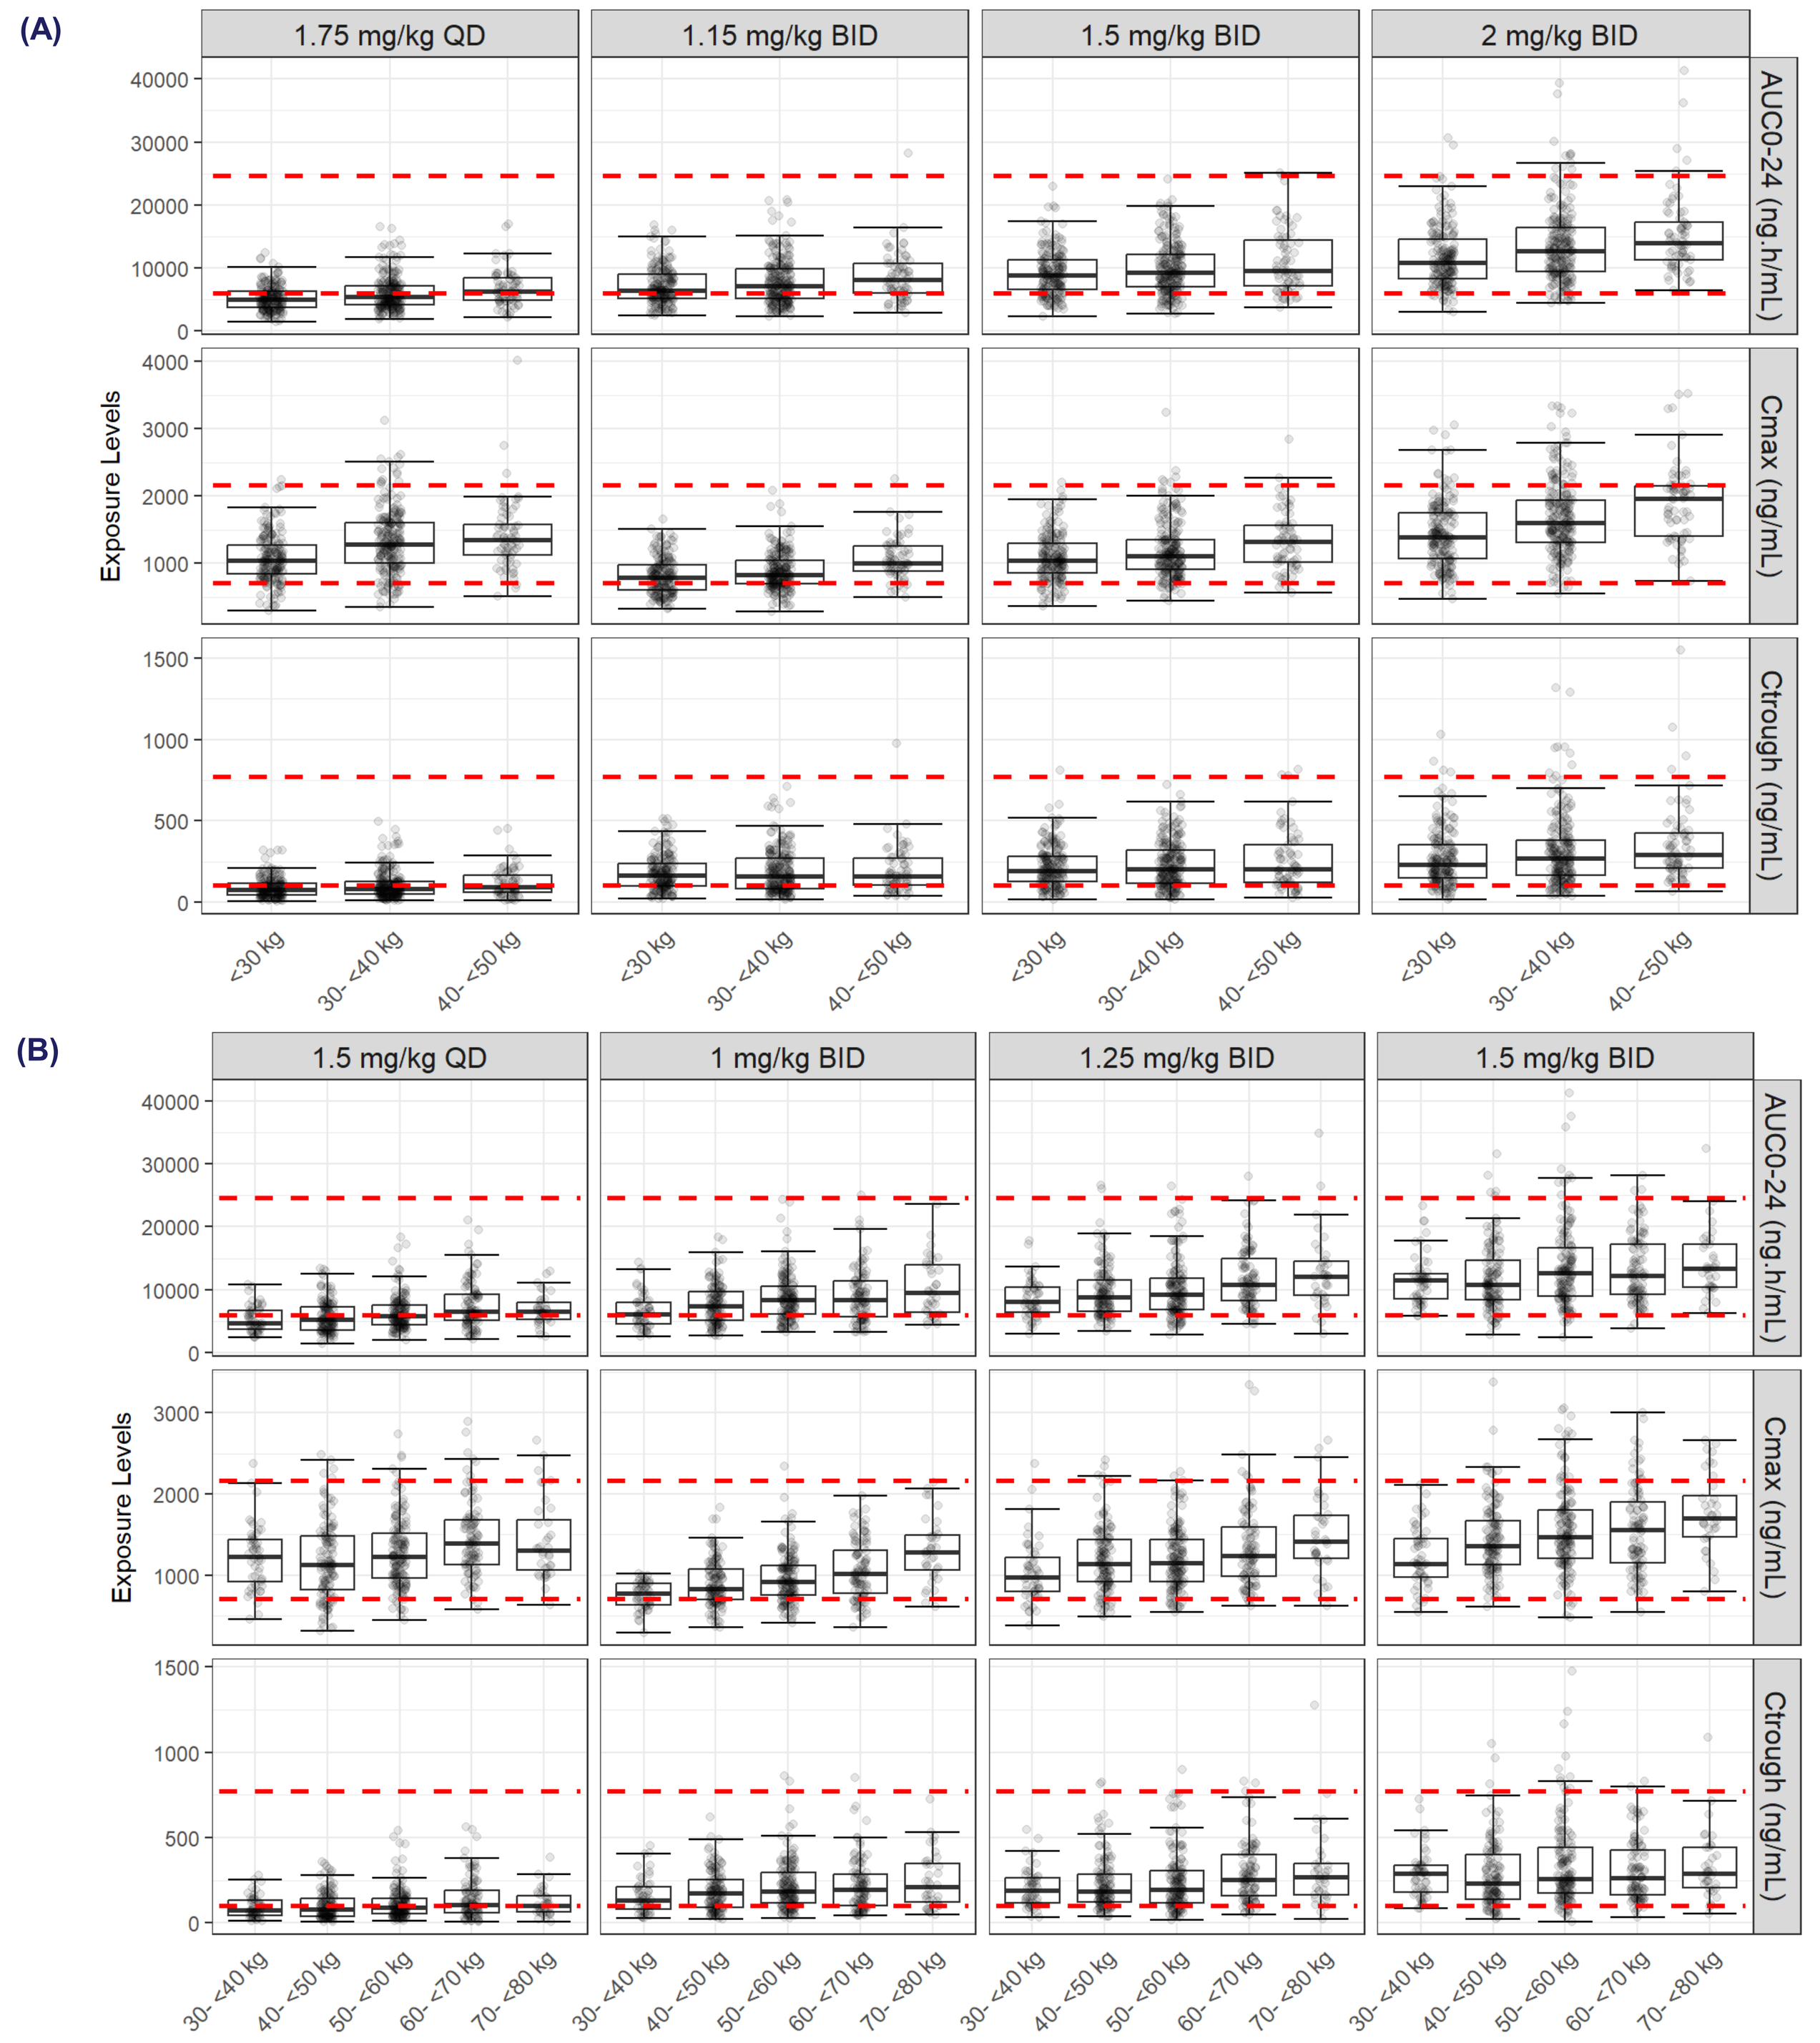

(A)
1.75 mg/kg QD
1.15 mg/kg BID
1.5 mg/kg BID
2 mg/kg BID
AUC0-24 (ng.h/mL)
Cmax (ng/mL)
Ctrough (ng/mL)
Exposure Levels
<30 kg
30-<40 kg
40-<50 kg
(B)
1.5 mg/kg QD
1 mg/kg BID
1.25 mg/kg BID
1.5 mg/kg BID
30-<40 kg
40-<50 kg
50-<60 kg
60-<70 kg
70-<80 kg

Abbreviations: AUC_{0-24}, area under the curve from time 0 to 24 hours, BID, bis in die (twice daily), cIAI, complicated intra-abdominal infection; C_{max}, maximum plasma concentration; C_{trough}, trough concentration; QD, quaque die (once daily);
Note: Only weight groups with N>20 are displayed; Red dashed lines represent the 5th and 95th percentiles of exposure in adults at 1 mg/kg BID.

We are grateful to the patients and their families for their participation, trust, and commitment. We also extend our sincere thanks to the site staff and CRO partners for their dedication and professionalism in conducting the trials.

clinicaltrials.gov

Figure 1. Visual Predictive Check – Model-Predicted and Observed Concentrations of Eravacycline Linear (A) and Semi-Log (B) Scales

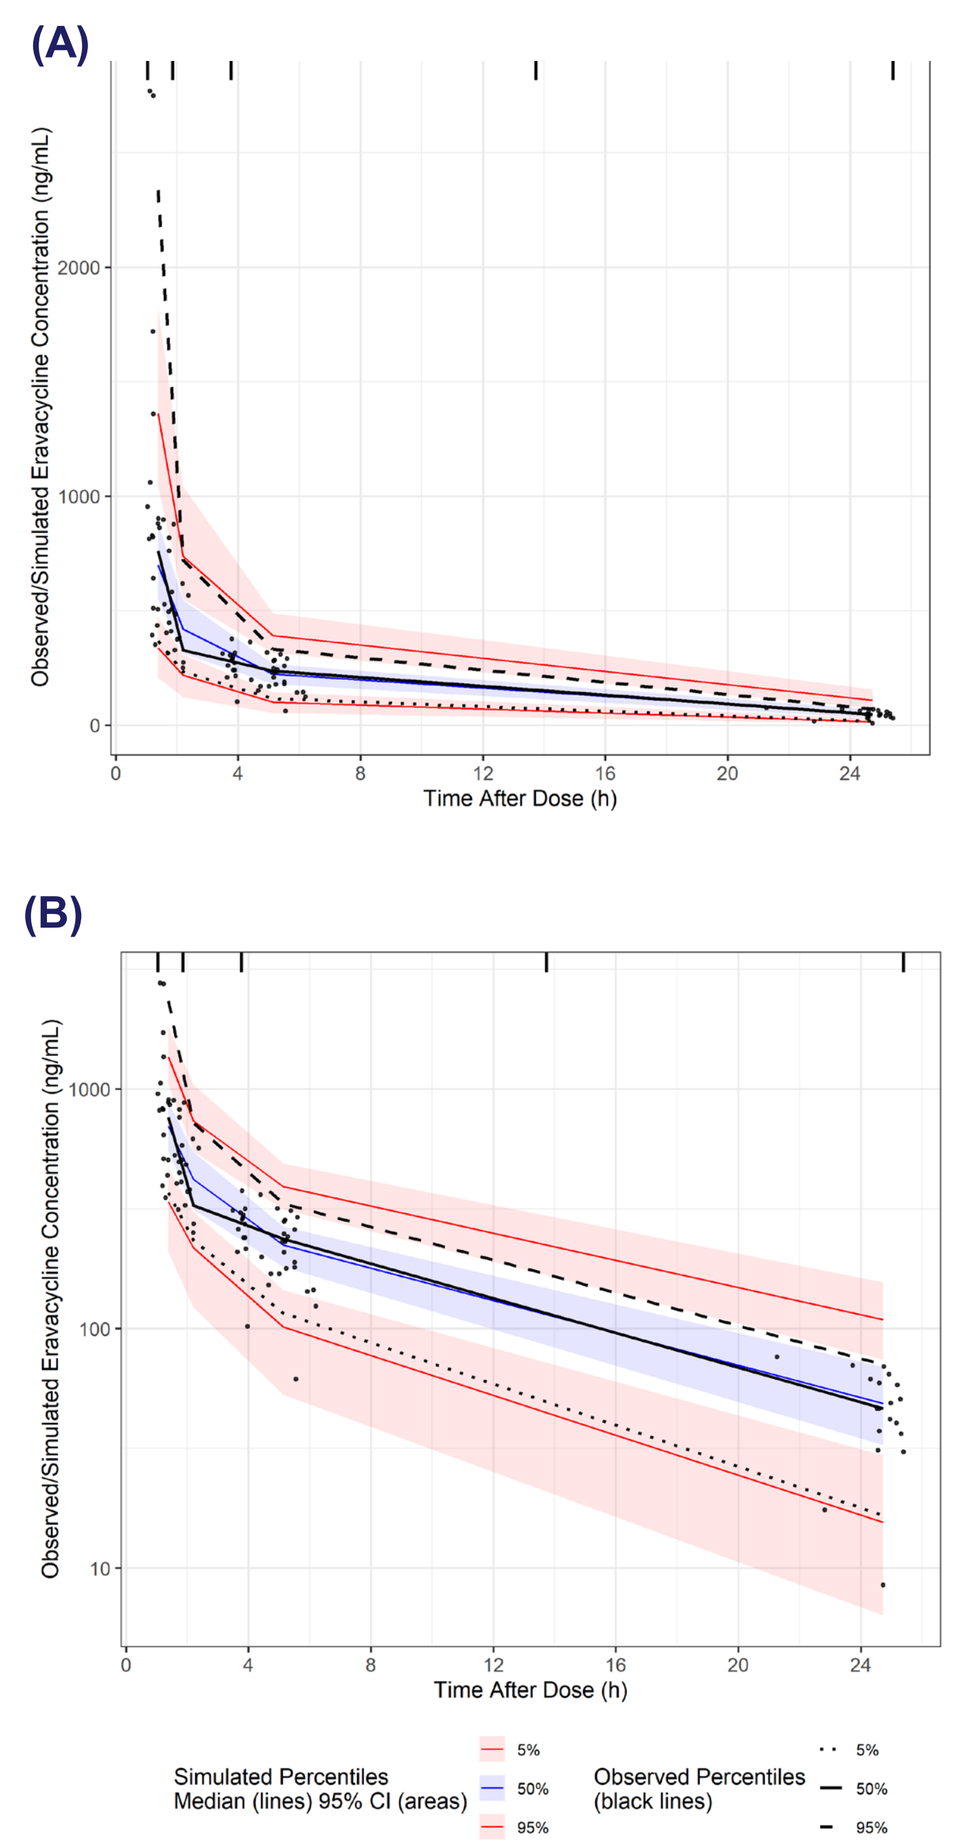

(A)
Observed/Simulated Eravacycline Concentration (ng/mL)
Time After Dose (h)
(B)
Simulated Percentiles
Median (lines) 95% CI (areas)
Observed Percentiles
(black lines)
5%
50%
95%

Table 3. Typical Population PK Parameters of Eravacycline Following Eravacycline IV Administration – Adults, Children and Adolescents

Parameter	Estimate (RSE)	BSV (Shrinkage)
CL (L/h)	11.8 (2.1%) × $(Weight/70)^{0.679}$ × $(Age/45)^{-0.287}$ × 0.674 if severe HEP × 0.654 if strong CYP3A inhibitor × 1.72 if strong CYP3A4 inducer	43.6% (11.2%)
V1 (L)	22.5 (11.2%) × $(Weight/70)^{0.421}$ ×2.14 if cIAI × 1.67 if cUTI × 3.39 if other bacterial infection	71.3% (15.7%)
Q2 (L/h)	32.1 (3.3%) × $(Weight/70)^{0.679}$ × exp(-0.305 × (Dose/Weight – 1.0))	51.8% (24.6%)
V2 (L)	109 (2.5%) × $(Weight/70)^{0.421}$	36.0% (31.5%)
Q3 (L/h)	5.25 (3.4%) × $(Weight/70)^{0.679}$ × exp(-0.285 × (Dose/Weight – 1.0))	
V3 (L)	175 (1.7%) × $(Weight/70)^{0.421}$	
Proportional Error	19.3%	
Additive Error (ng/mL)	1.54	

Abbreviations: BSV, between subject variability; cIAI, complicated intra-abdominal infection; CL, clearance; cUTI, complicated urinary tract infection; CYP3A, cytochrome P450 3A; HEP, hepatic impairment; IV, intravenous; PK, pharmacokinetic; Q2 and Q3, intercompartmental clearances; RSE, relative standard error; V1, central volume of distribution; V2 and V3, peripheral volumes of distribution
Note: Correction factors of 1.79 and 1.76 were applied for the different assays used in two studies

Table 4. Summary of Predicted Steady-State Exposures in Children, Adolescents, and Adults with cIAI

	Pediatric Patients		Adult Patients
	Children (8 years to <12 years)	Adolescents (12 years to <18 years)	
	2 mg/kg q12h (N=500)	1.5 mg/kg q12h (N=500)	1 mg/kg q12h (N=500)
Age (years) **Mean [Min, Max]**	9.93 [8.04, 12.0]	15.1 [12.0, 18.0]	52.6 [18.0, 88.0]
Weight **Mean [Min, Max]**	33.5 [19.5, 67.6]	56.0 [29.2, 134]	77.9 [32.0, 137]
Total daily dose (mg) **Mean [Min, Max]**	134 [78.0, 270]	168 [87.6, 401]	156 [64.0, 274]
C_{max} (ng/mL) **Median [Min, Max]** **Geometric Mean (%CV)**	1529 [471, 4014] 1525 (36.7%)	1456 [485, 3381] 1448 (34.8%)	1258 [416, 2994] 1266 (34.1%)
AUC_{0-24} (h•ng/mL) **Median [Min, Max]** **Geometric Mean (%CV)**	12034 [2958, 41423] 11985 (43.6%)	11874 [2366, 41309] 11887 (45.0%)	12333 [3357, 34768] 12146 (44.0%)

Abbreviations: %CV, percent coefficient of variation; AUC_{0-24}, area under the curve from time 0 to 24 hours, cIAI, complicated intra-abdominal infection; C_{max}, maximum plasma concentration; max, maximum; min, minimum; PK, pharmacokinetic; q12h, every 12 hours
Note, AUC_{0-24} is calculated as AUC_{0-12} x 2

Conclusions

- The following ERV doses are anticipated to be safe and efficacious in pediatric patients (8 to < 18 years of age) with cIAI and were derived from this updated population PK model:
 - 8 to < 12 years at a dose of 2 mg/kg q12h
 - 12 years to < 18 years at a dose of 1.5 mg/kg q12h
- These doses are being evaluated in a Phase 2 pediatric clinical trial (NCT06794541)
- PK and safety data will be reviewed during interim analyses to evaluate the appropriateness of the doses

Presented at IDWeek 2025, Atlanta, GA